When recognition is dominated by regularity: Perceptual and lexical decisions under semantic impairment

Timothy T. Rogers, Matthew A. Lambon Ralph*, John R. Hodges, Karalyn Patterson

INTRODUCTION

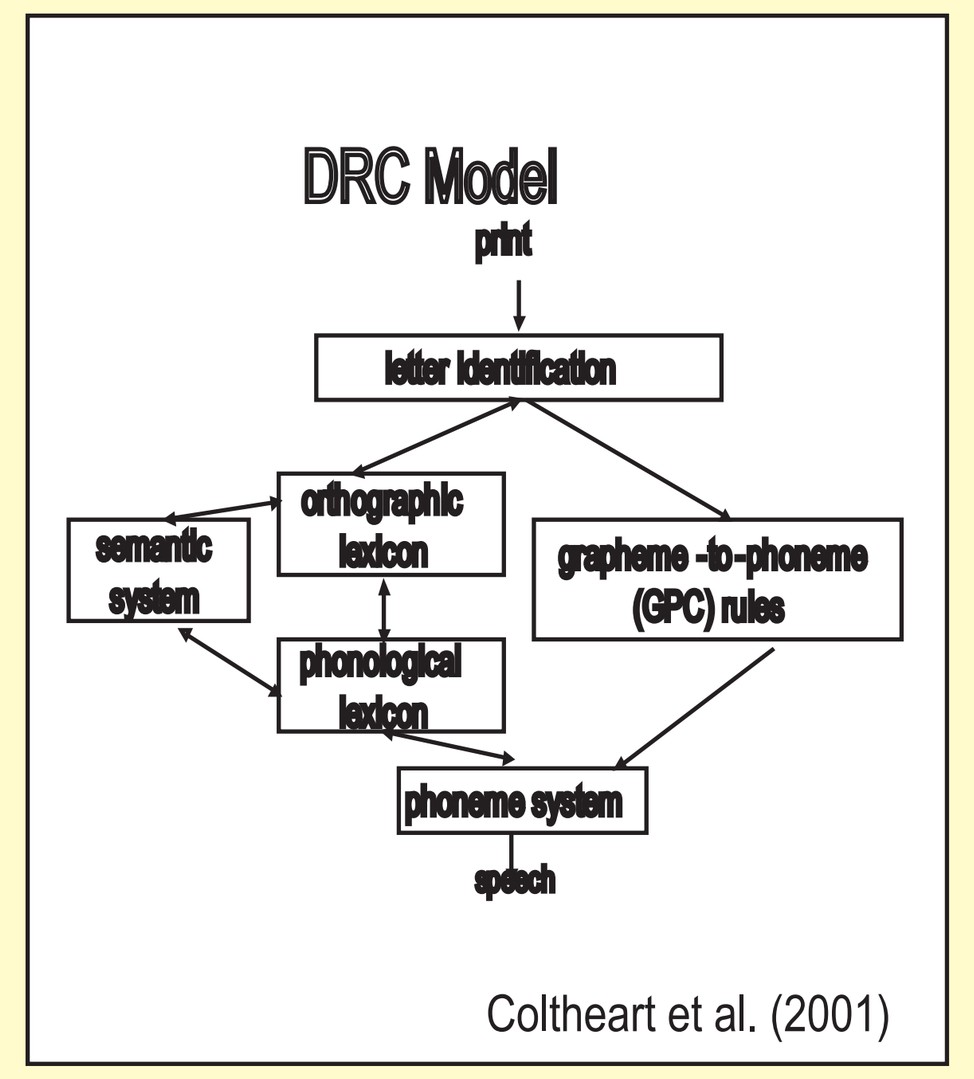
DRC Model
print
letter identification
orthographic lexicon
semantic system
grapheme -to-phoneme (GPC) rules
phonological lexicon
phoneme system
speech
Coltheart et al. (2001)

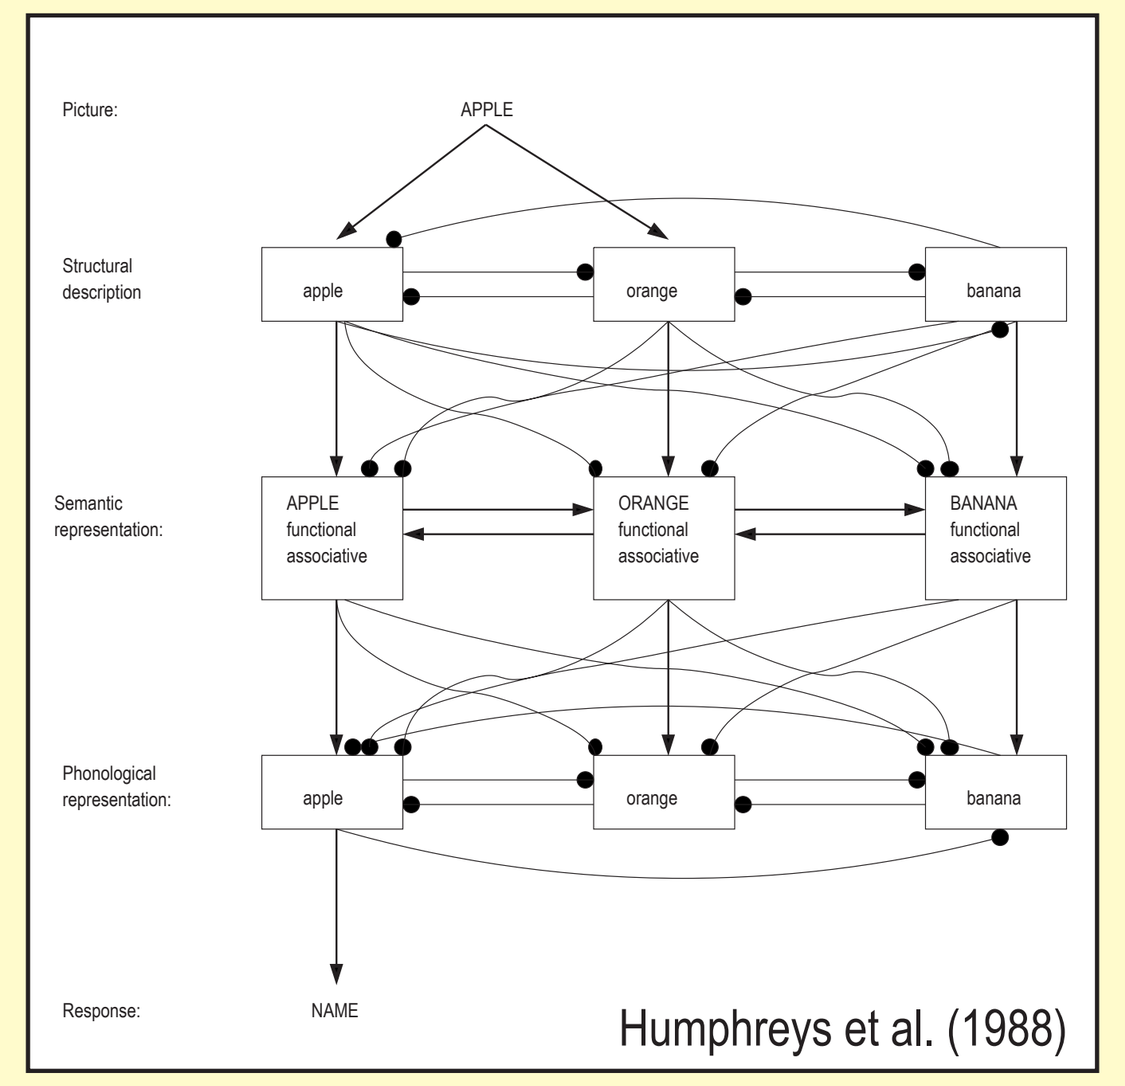
Picture:
APPLE
Structural description:
apple
orange
banana
Semantic representation:
APPLE functional associative
ORANGE functional associative
BANANA functional associative
Phonological representation:
Response:
NAME
Humphreys et al. (1988)

■ Models of written-word recognition often invoke pre-semantic lexical representations that are capable of mediating word-recognition without support from semantics. For example, the dual-route cascaded model posits an orthographic lexicon, which provides input to semantics from orthography, but which allows written-word recognition independent of

■ Similarly, many models of visual object recognition posit that visual stimuli first activate stored pre-semantic structural descriptions that mediate recognition, which feed forward to activate semantic representations. Visual recognition can be spared under semantic impairment because semantic processing does not influence the activation of visual structural descriptions.

Alternatively...

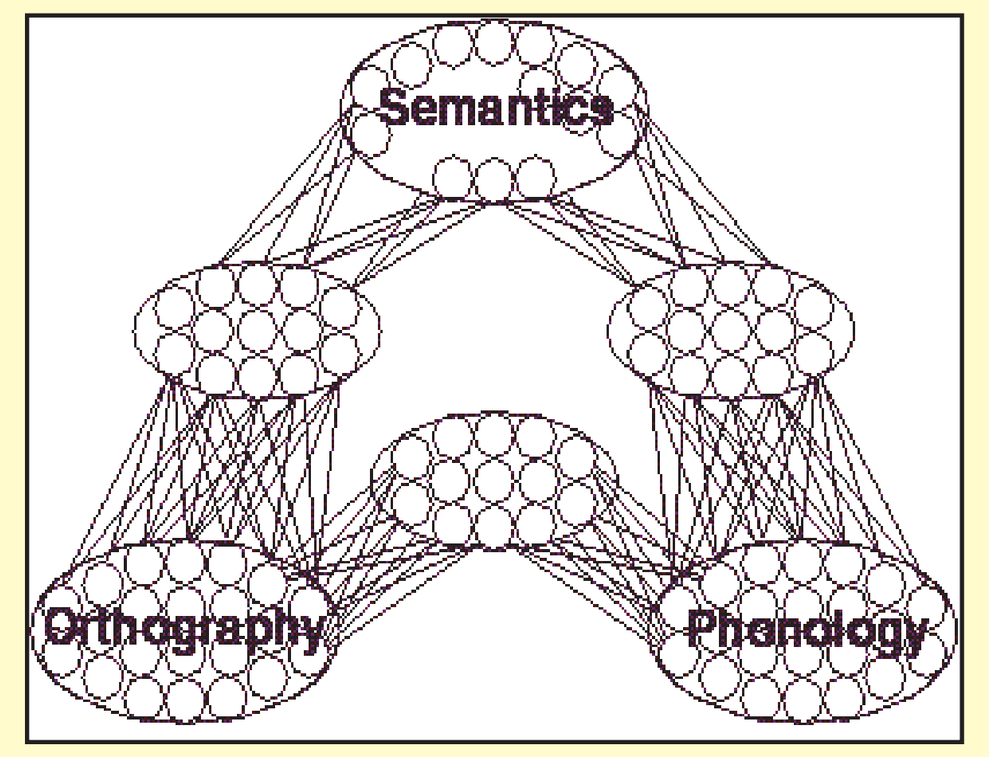
Semantics
Orthography
Phonology
Plaut et al. (1996)

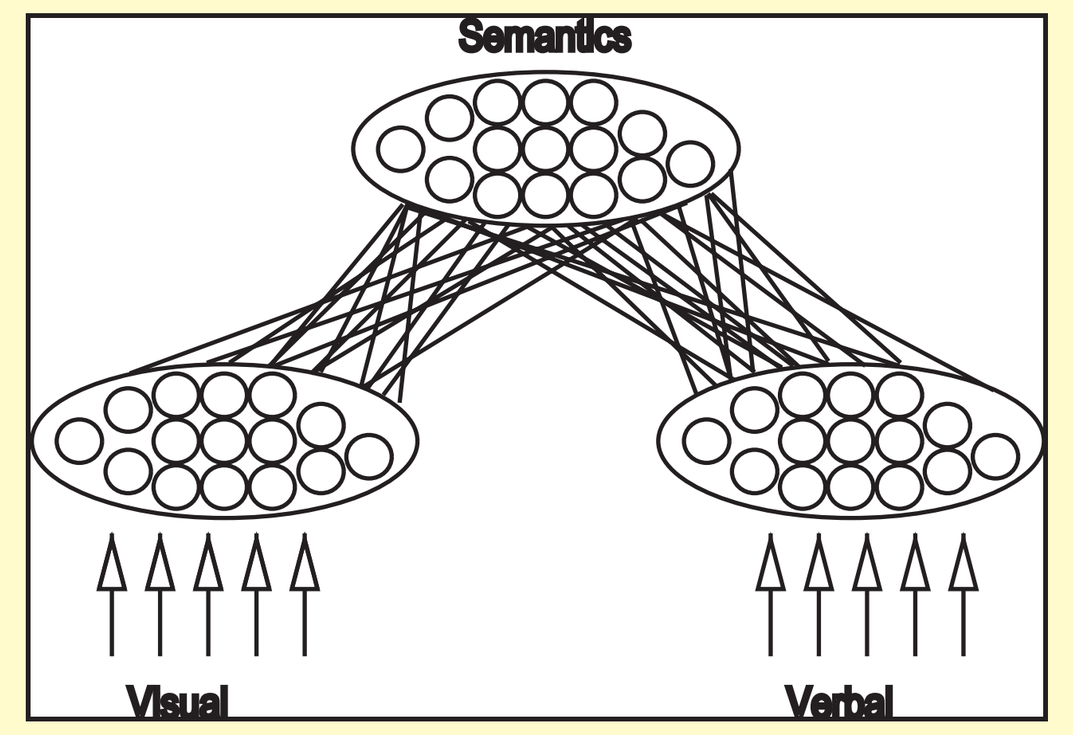
Semantics
Visual
Verbal
Rogers et al. (1999)

■ Alternatively, distributed connectionist models of both written-word and visual-semantic processing suggest that distributed representations at multiple levels interact and mutually support one another via semantics. Disruptions anywhere in the system can affect processing, but the particular effects observed depend upon the structure of the representations at each level. Irregular items, such as words with unusual orthographies or objects with atypical appearances, rely to a greater extent on the engagement of semantic representations for successful recognition.

■ In the current work we tested the effects of semantic impairment on visual object and written-word recognition, in tasks that varied the regularity of the target and distractor items. Models that invoke pre-semantic representations to support recognition predict either spared recognition with semantic impairment, or impaired recognition for both regular and irregular items. Distributed connectionist accounts predict increasingly impaired recognition for irregular items as semantic knowledge degrades, with relative sparing of recognition for regular items.

EXPERIMENT 1: OVER-REGULAR ANIMAL TEST (OAT)

■ Task: Which one is real?
■ For non-real (NR) > real (R) stimuli, chimera is more prototypical than target (like gorillas on right).
■ For R > NR stimuli, chimera is less typical than target (like lions on right).
■ NR > R and R > NR item pairs matched for difference between target and chimera. In examples shown, both items include an animal with a tail and an animal without a tail.
■ 16 items in each condition
■ 20 patients with semantic dementia tested

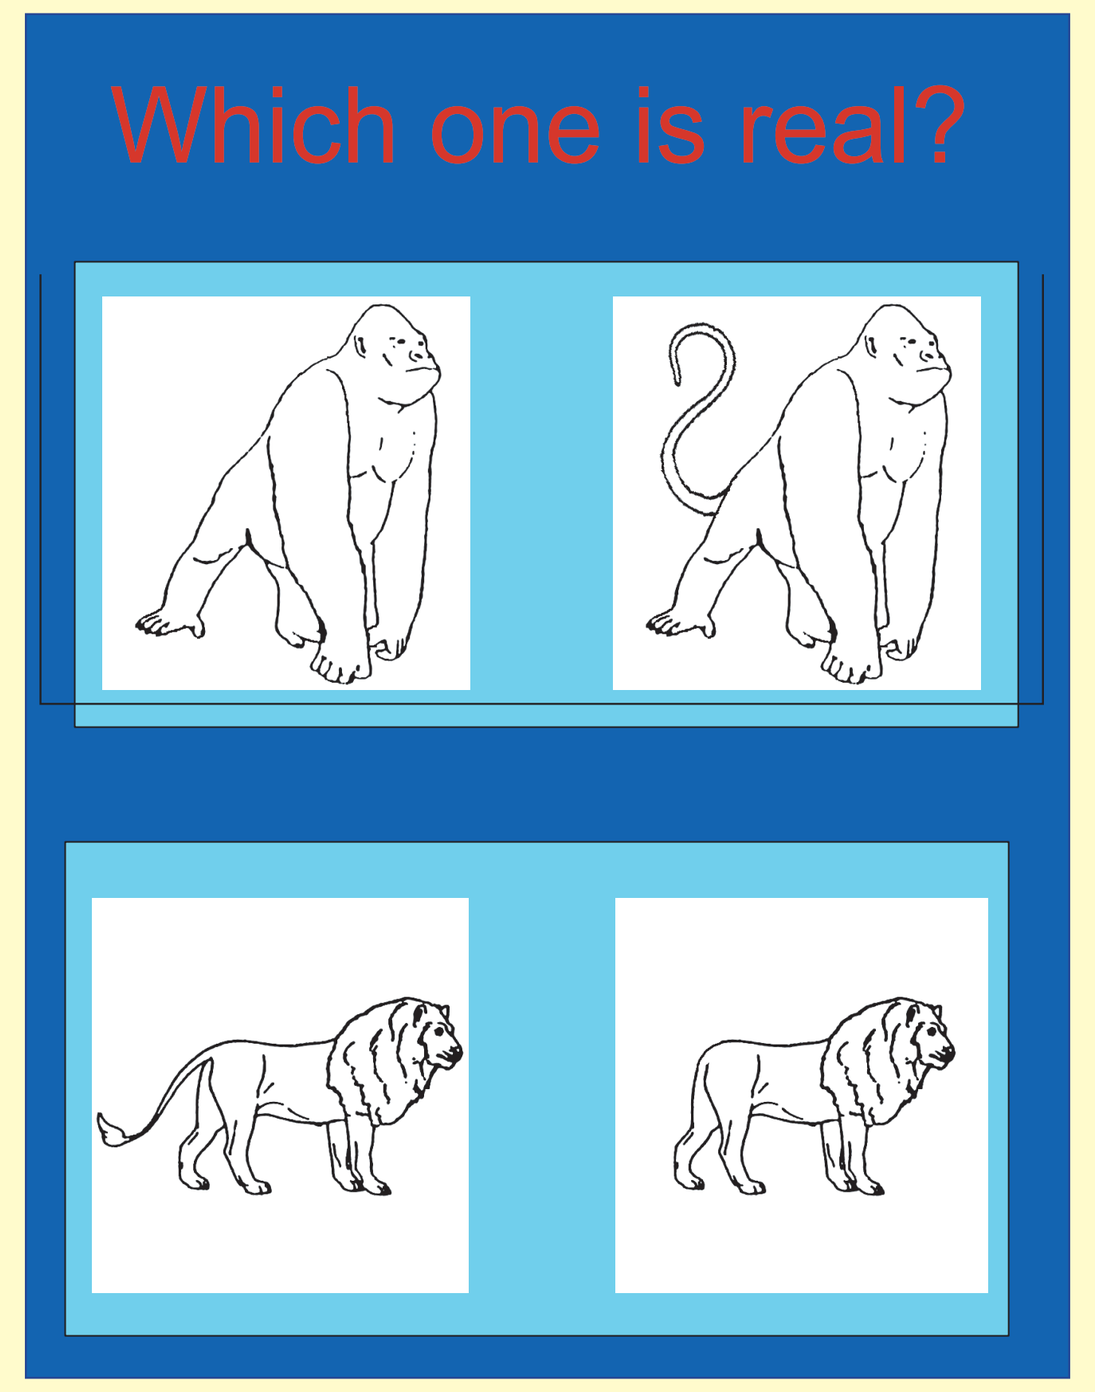
Which one is real?

Proportion correct by condition and patient

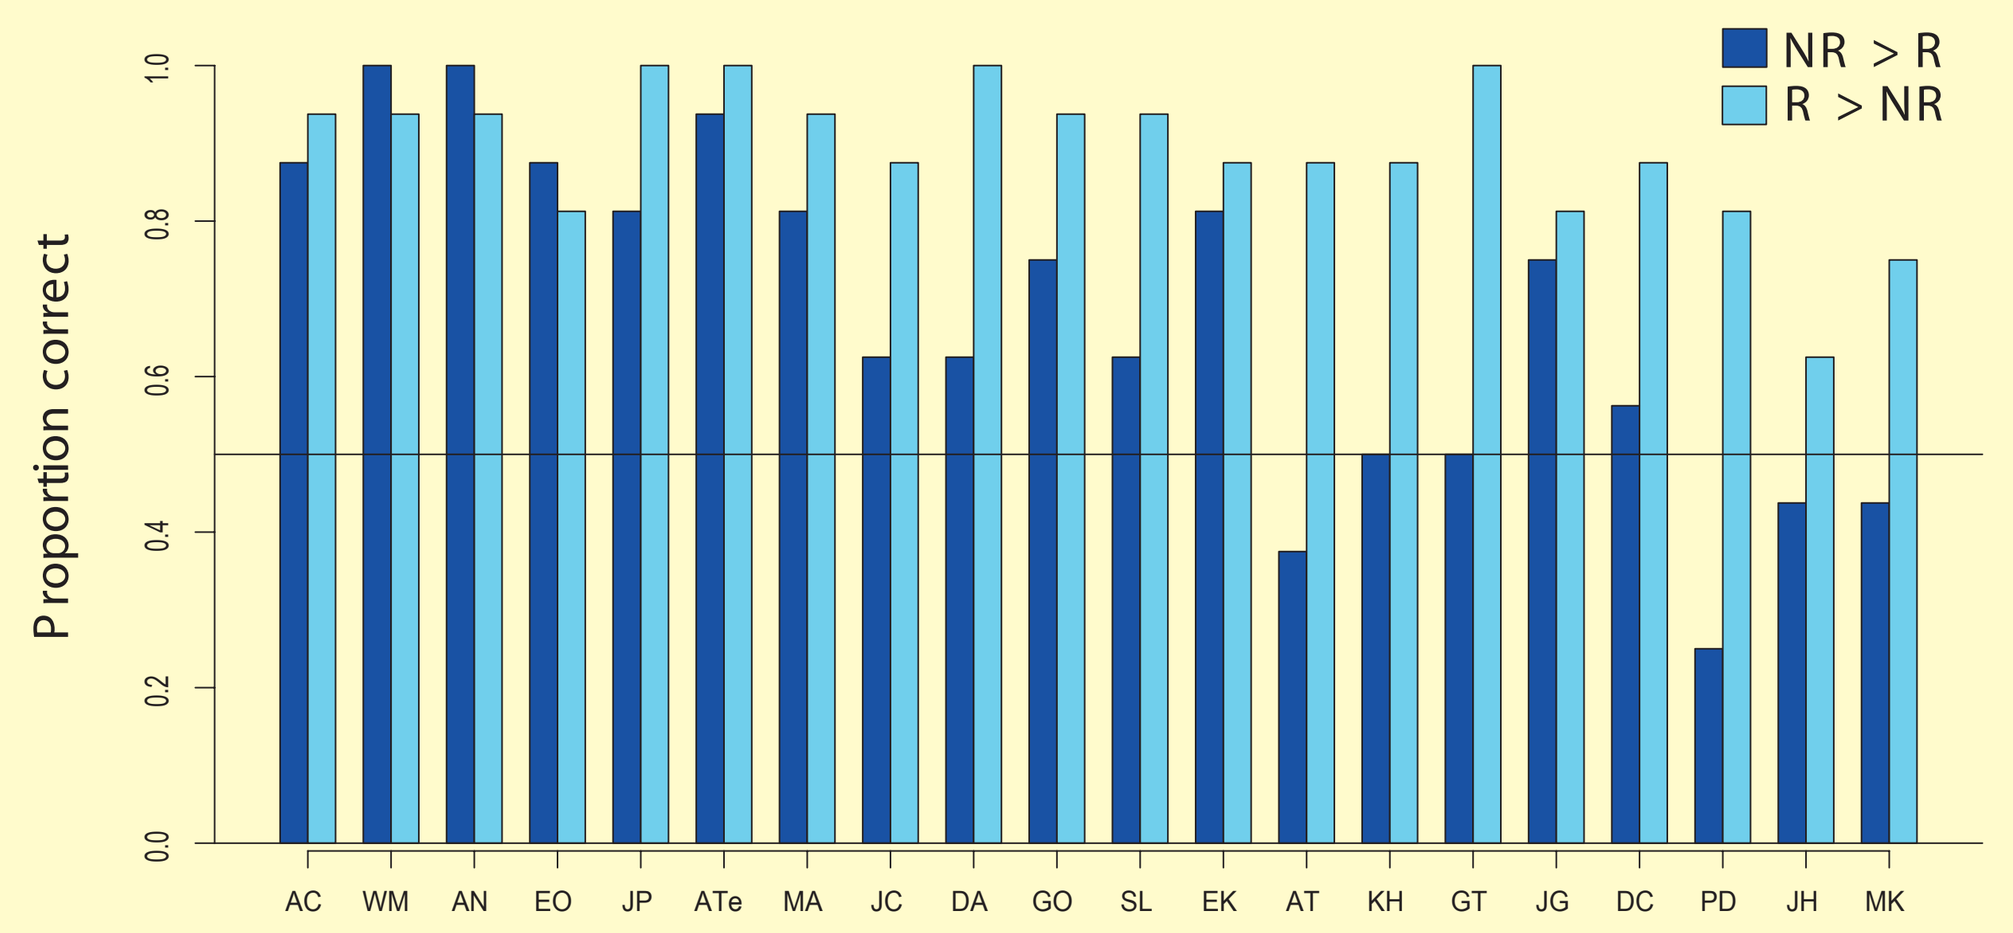
NR > R
R > NR
Proportion correct
AC WM AN EO JP ATe MA JC DA GO SL EK AT KH GT JG DC PD JH MK

■ Performance in both conditions deteriorates with magnitude of semantic impairment...
■ BUT performance is much worse when targets are unusual and distractors are regular (NR > R condition).

Comparison to BORB for 13 patients

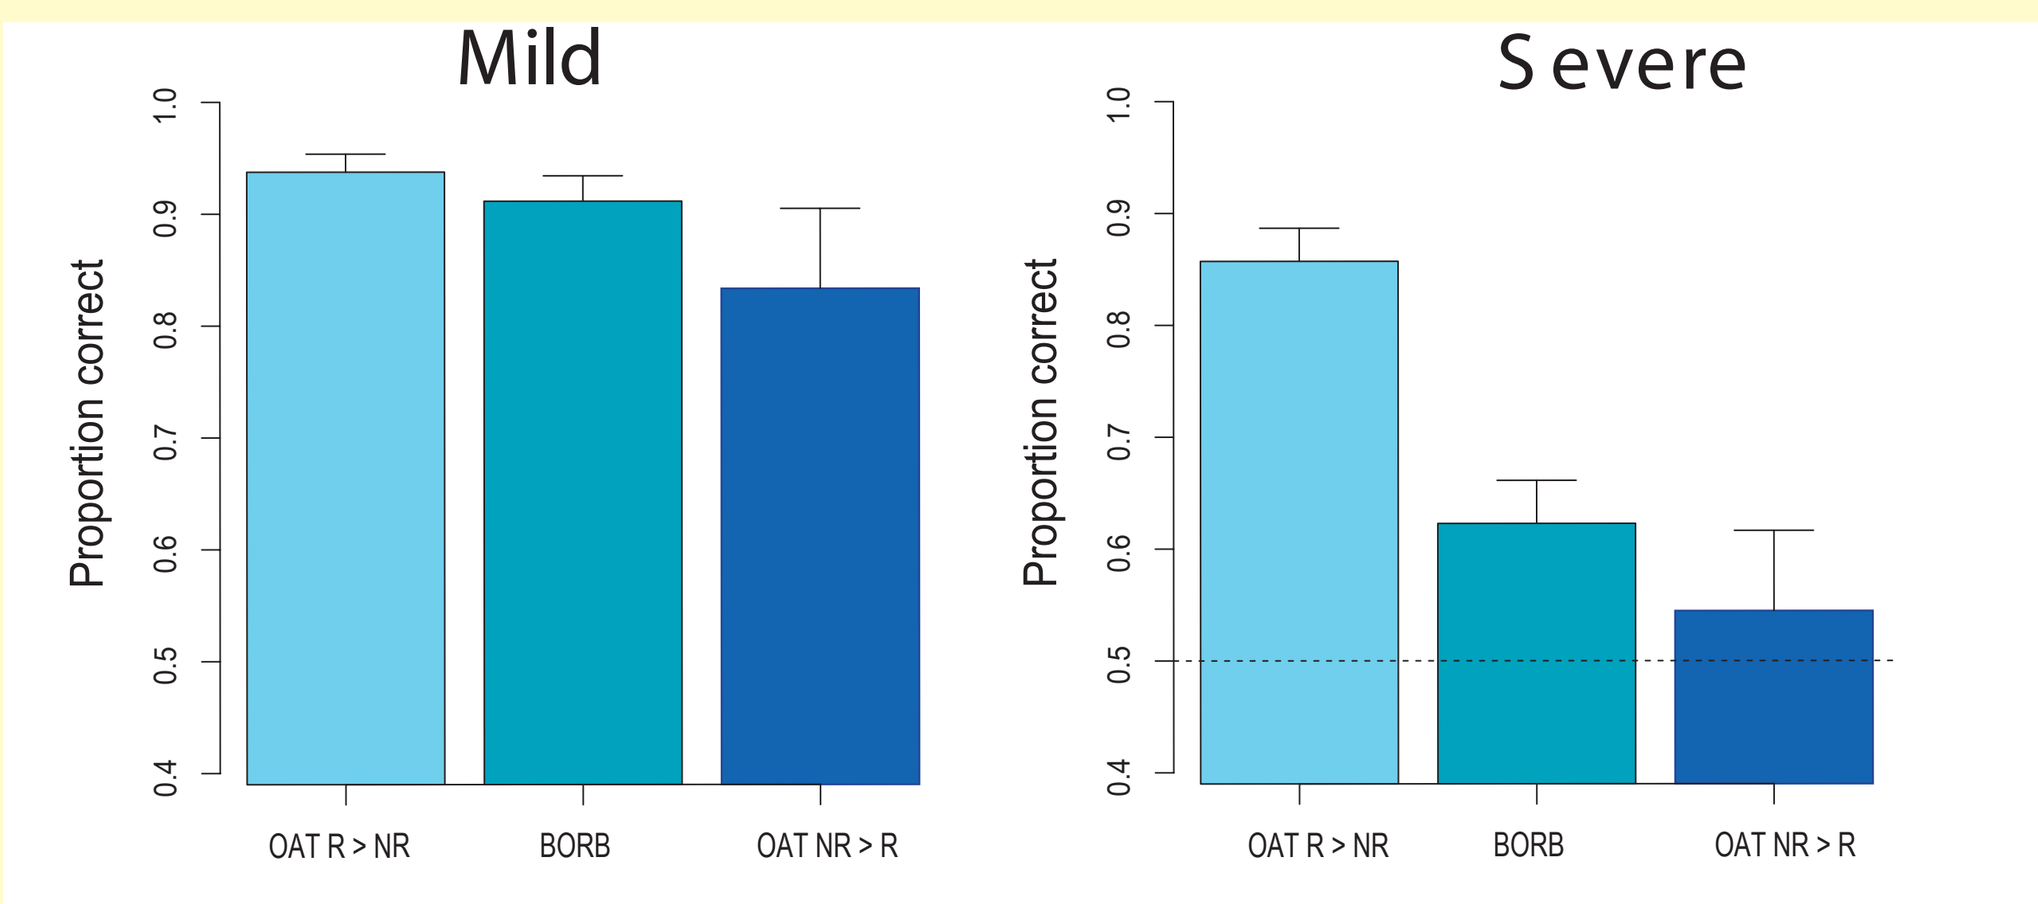
Mild
Severe
Proportion correct
OAT R > NR
BORB
OAT NR > R

13 patients were also tested on the Birmingham Object Recognition Battery (BORB). Milder patients performed well in both conditions of the OAT and on the BORB. 95% confidence intervals for mean proportion correct overlap for all three conditions.

More severe patients performed well on the R > NR condition of the OAT and poorly in the BORB and in the NR > R condition of the OAT. Performance was reliably better than chance in the BORB, but not in the NR > R condition of the OAT.

Over-regularisation effects in the BORB

Four patients completed the entire BORB (128 items total). From this large battery we culled:

■ 11 typical-looking chimeras
■ 11 typical-looking real animals
■ 11 atypical-looking chimeras
■ 11 atypical-looking real animals

Performance across the four patients varied as a function of stimulus type (real or chimeric) and typicality. When stimuli were typical, patients accepted them as real. When stimuli were atypical, patients guessed randomly.

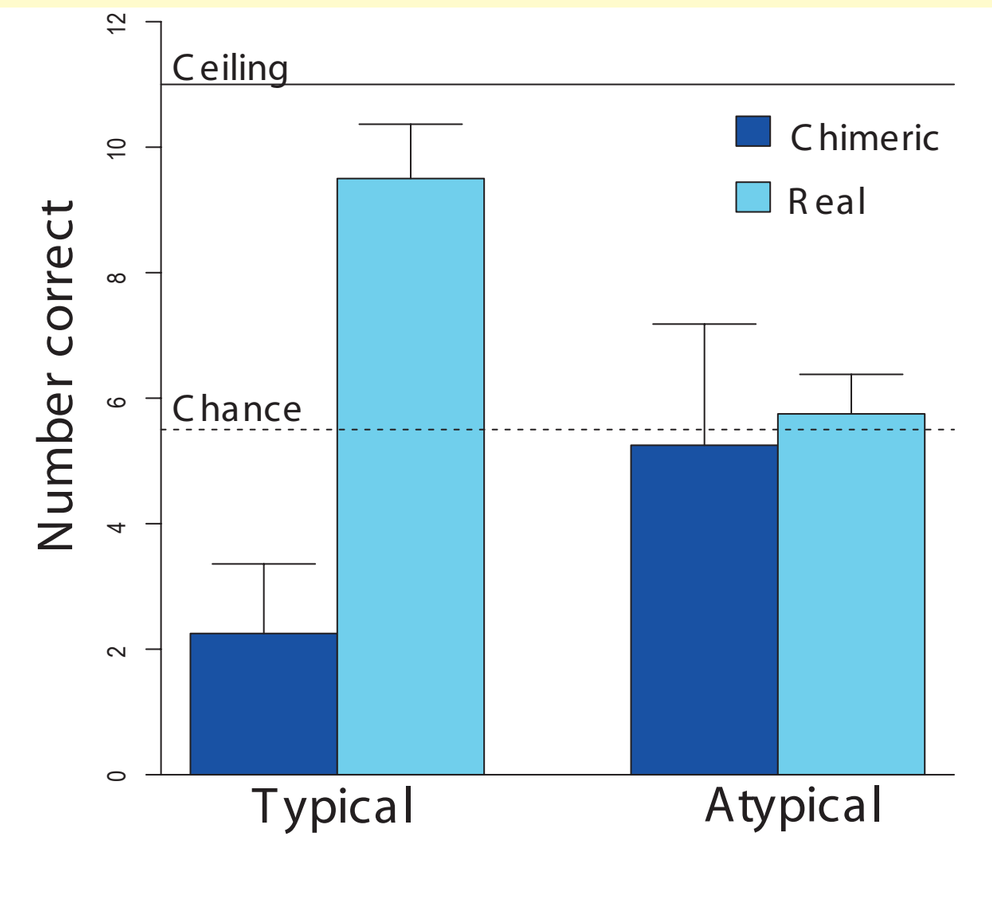
Ceiling
Chance
Chimeric
Real
Number correct
Typical
Atypical

EXPERIMENT 2: OVER-REGULAR WORD TEST (OWT)

Which one is real?

W > NW		NW > W	
LOT	LACHT	YOT	YACHT
GRIST	GRYST	TRIST	TRYST
DOLLOP	DOLYP	POLLOP	POLYP

■ 2-alternative forced-choice lexical decision
■ Real word and pseudo-nonword homophone appear side-by-side
■ Participant must choose which is the real word
■ For half of the items, nonword distractor is orthographically more "typical" than real word (NW > W condition)
■ For remaining items, reverse is true (W > NW condition).

Stimulus construction

■ 68 item pairs, 34 in each condition
■ Each NW>W item matched to an item in the W>NW condition for:
■ Frequency of target word
■ Orthographic difference between target and distractor

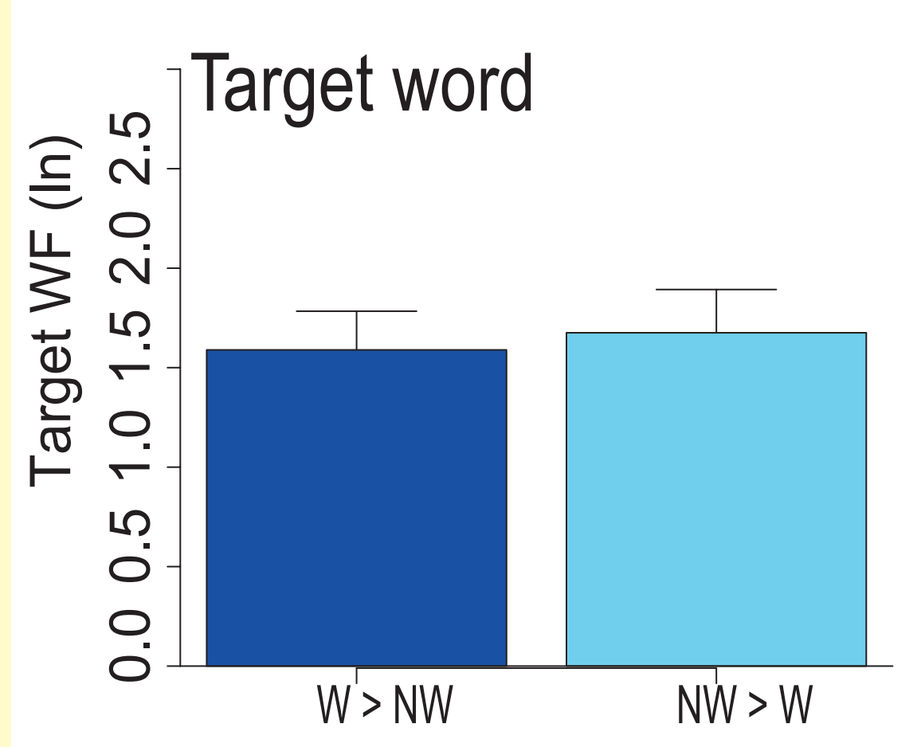
Target word
Target WF (ln)
W > NW
NW > W

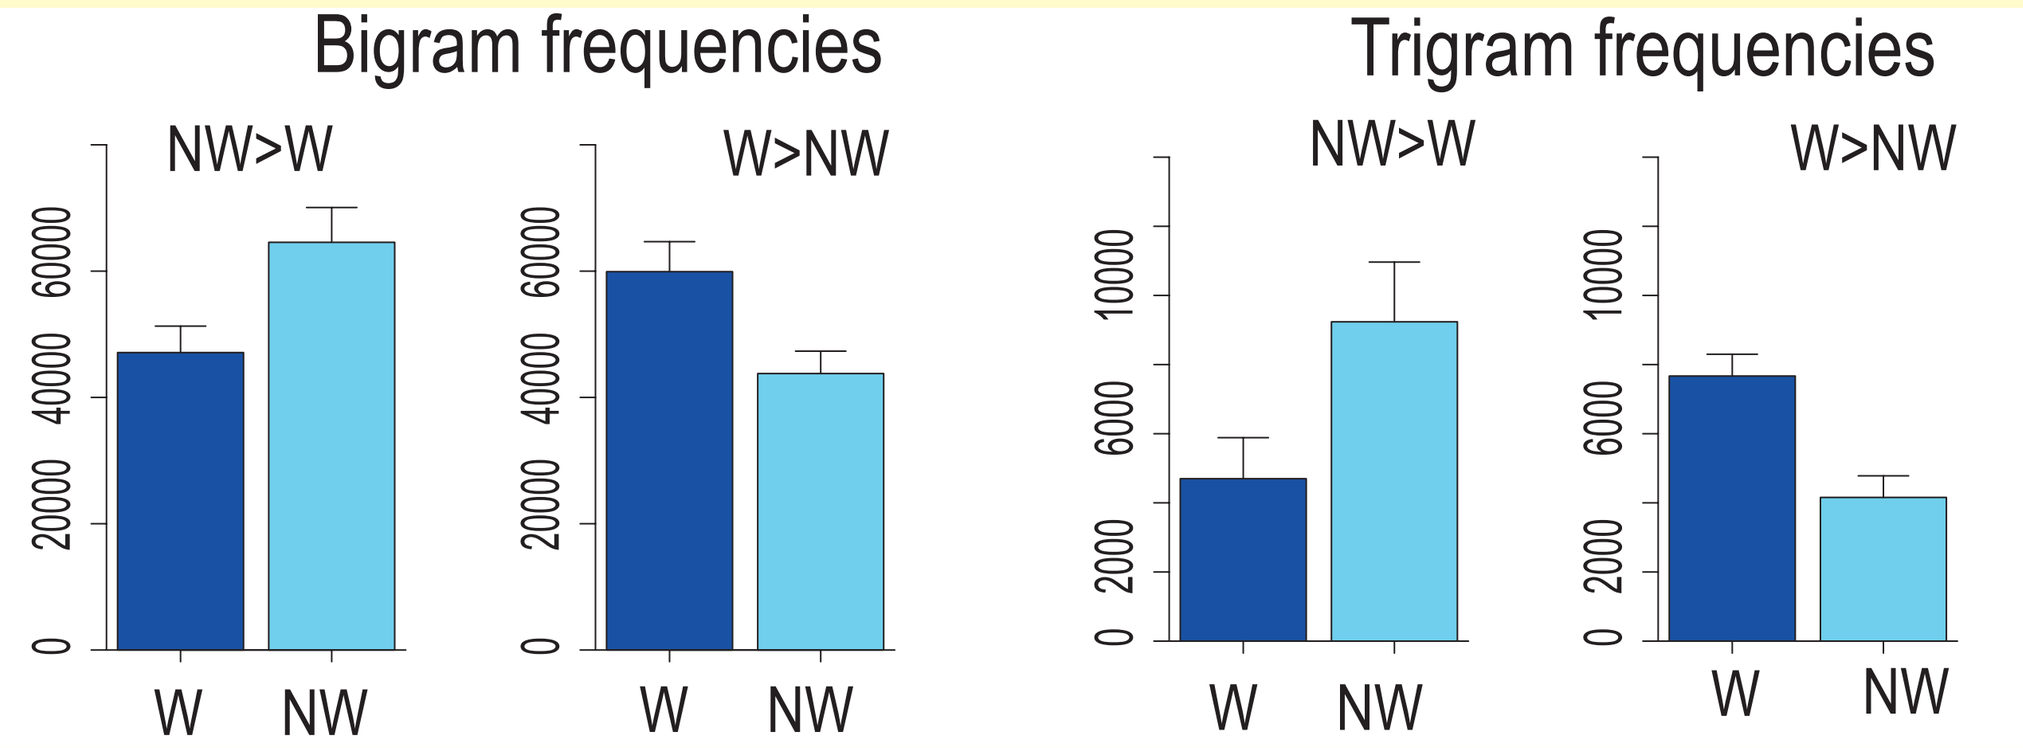
Bigram frequencies
Trigram frequencies
NW>W
W>NW
W
NW

Data collection

Data were collected from:
■ 13 patients with semantic dementia
■ 11 age- and education-matched controls
■ 5 patients with non-semantic language impairments (Broca's aphasia).

For each participant we examined:
■ Total proportion corect for W>NW and NW>W conditions
■ Proportion correct tabulated separately for low- and high-frequency target word pairs.

Control data

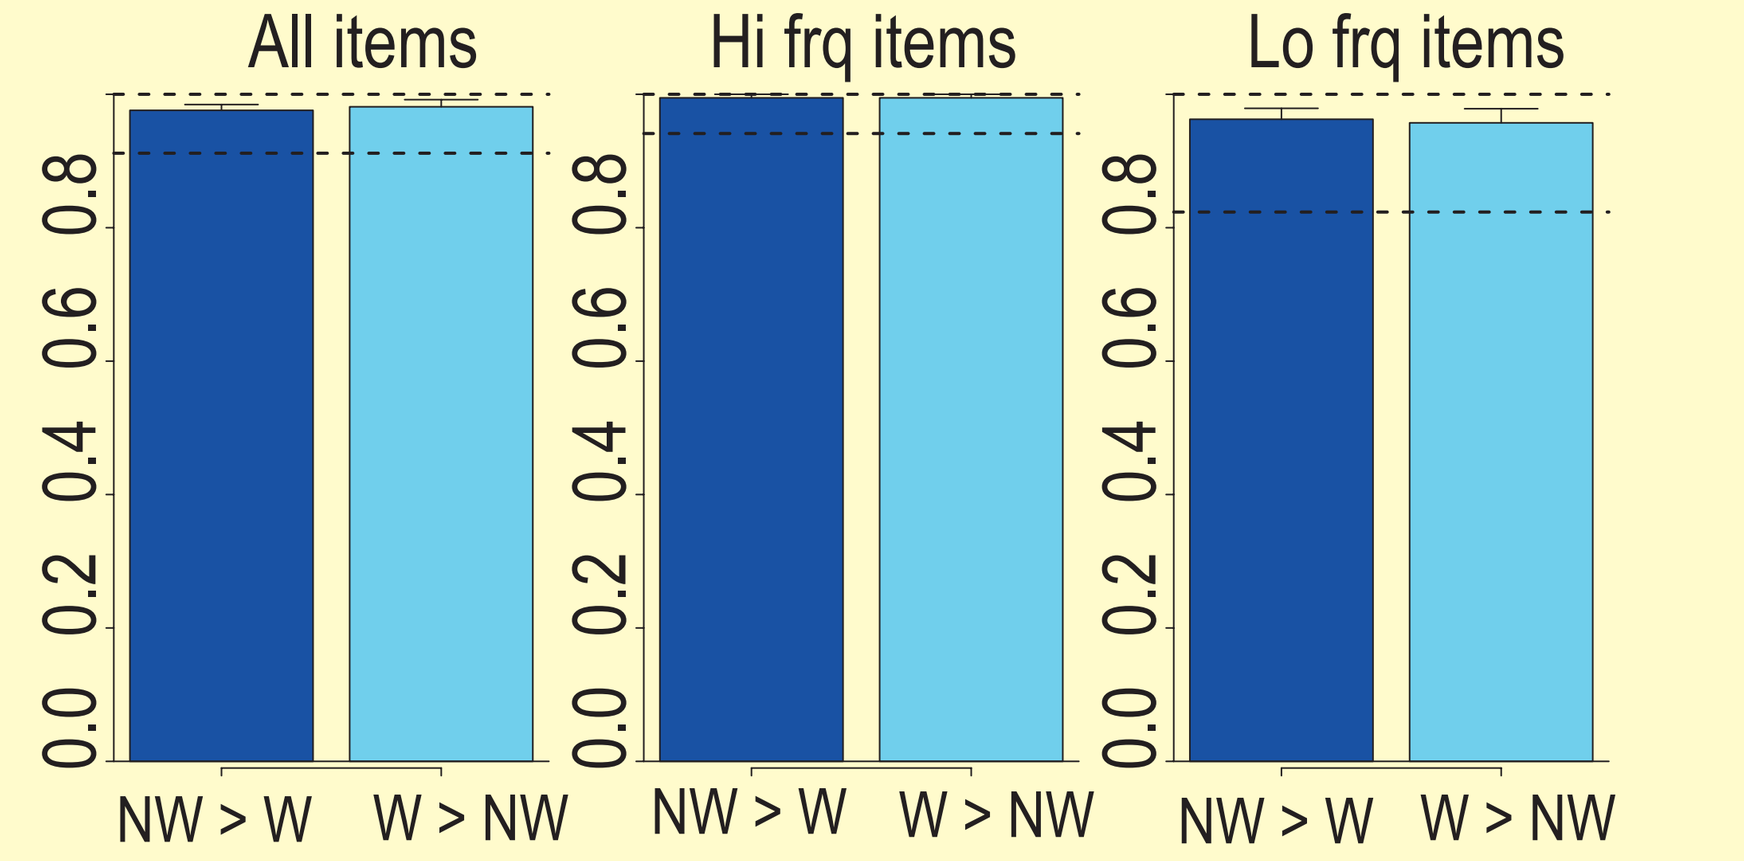
All items
Hi frq items
Lo frq items
NW > W
W > NW

Patient data: All items

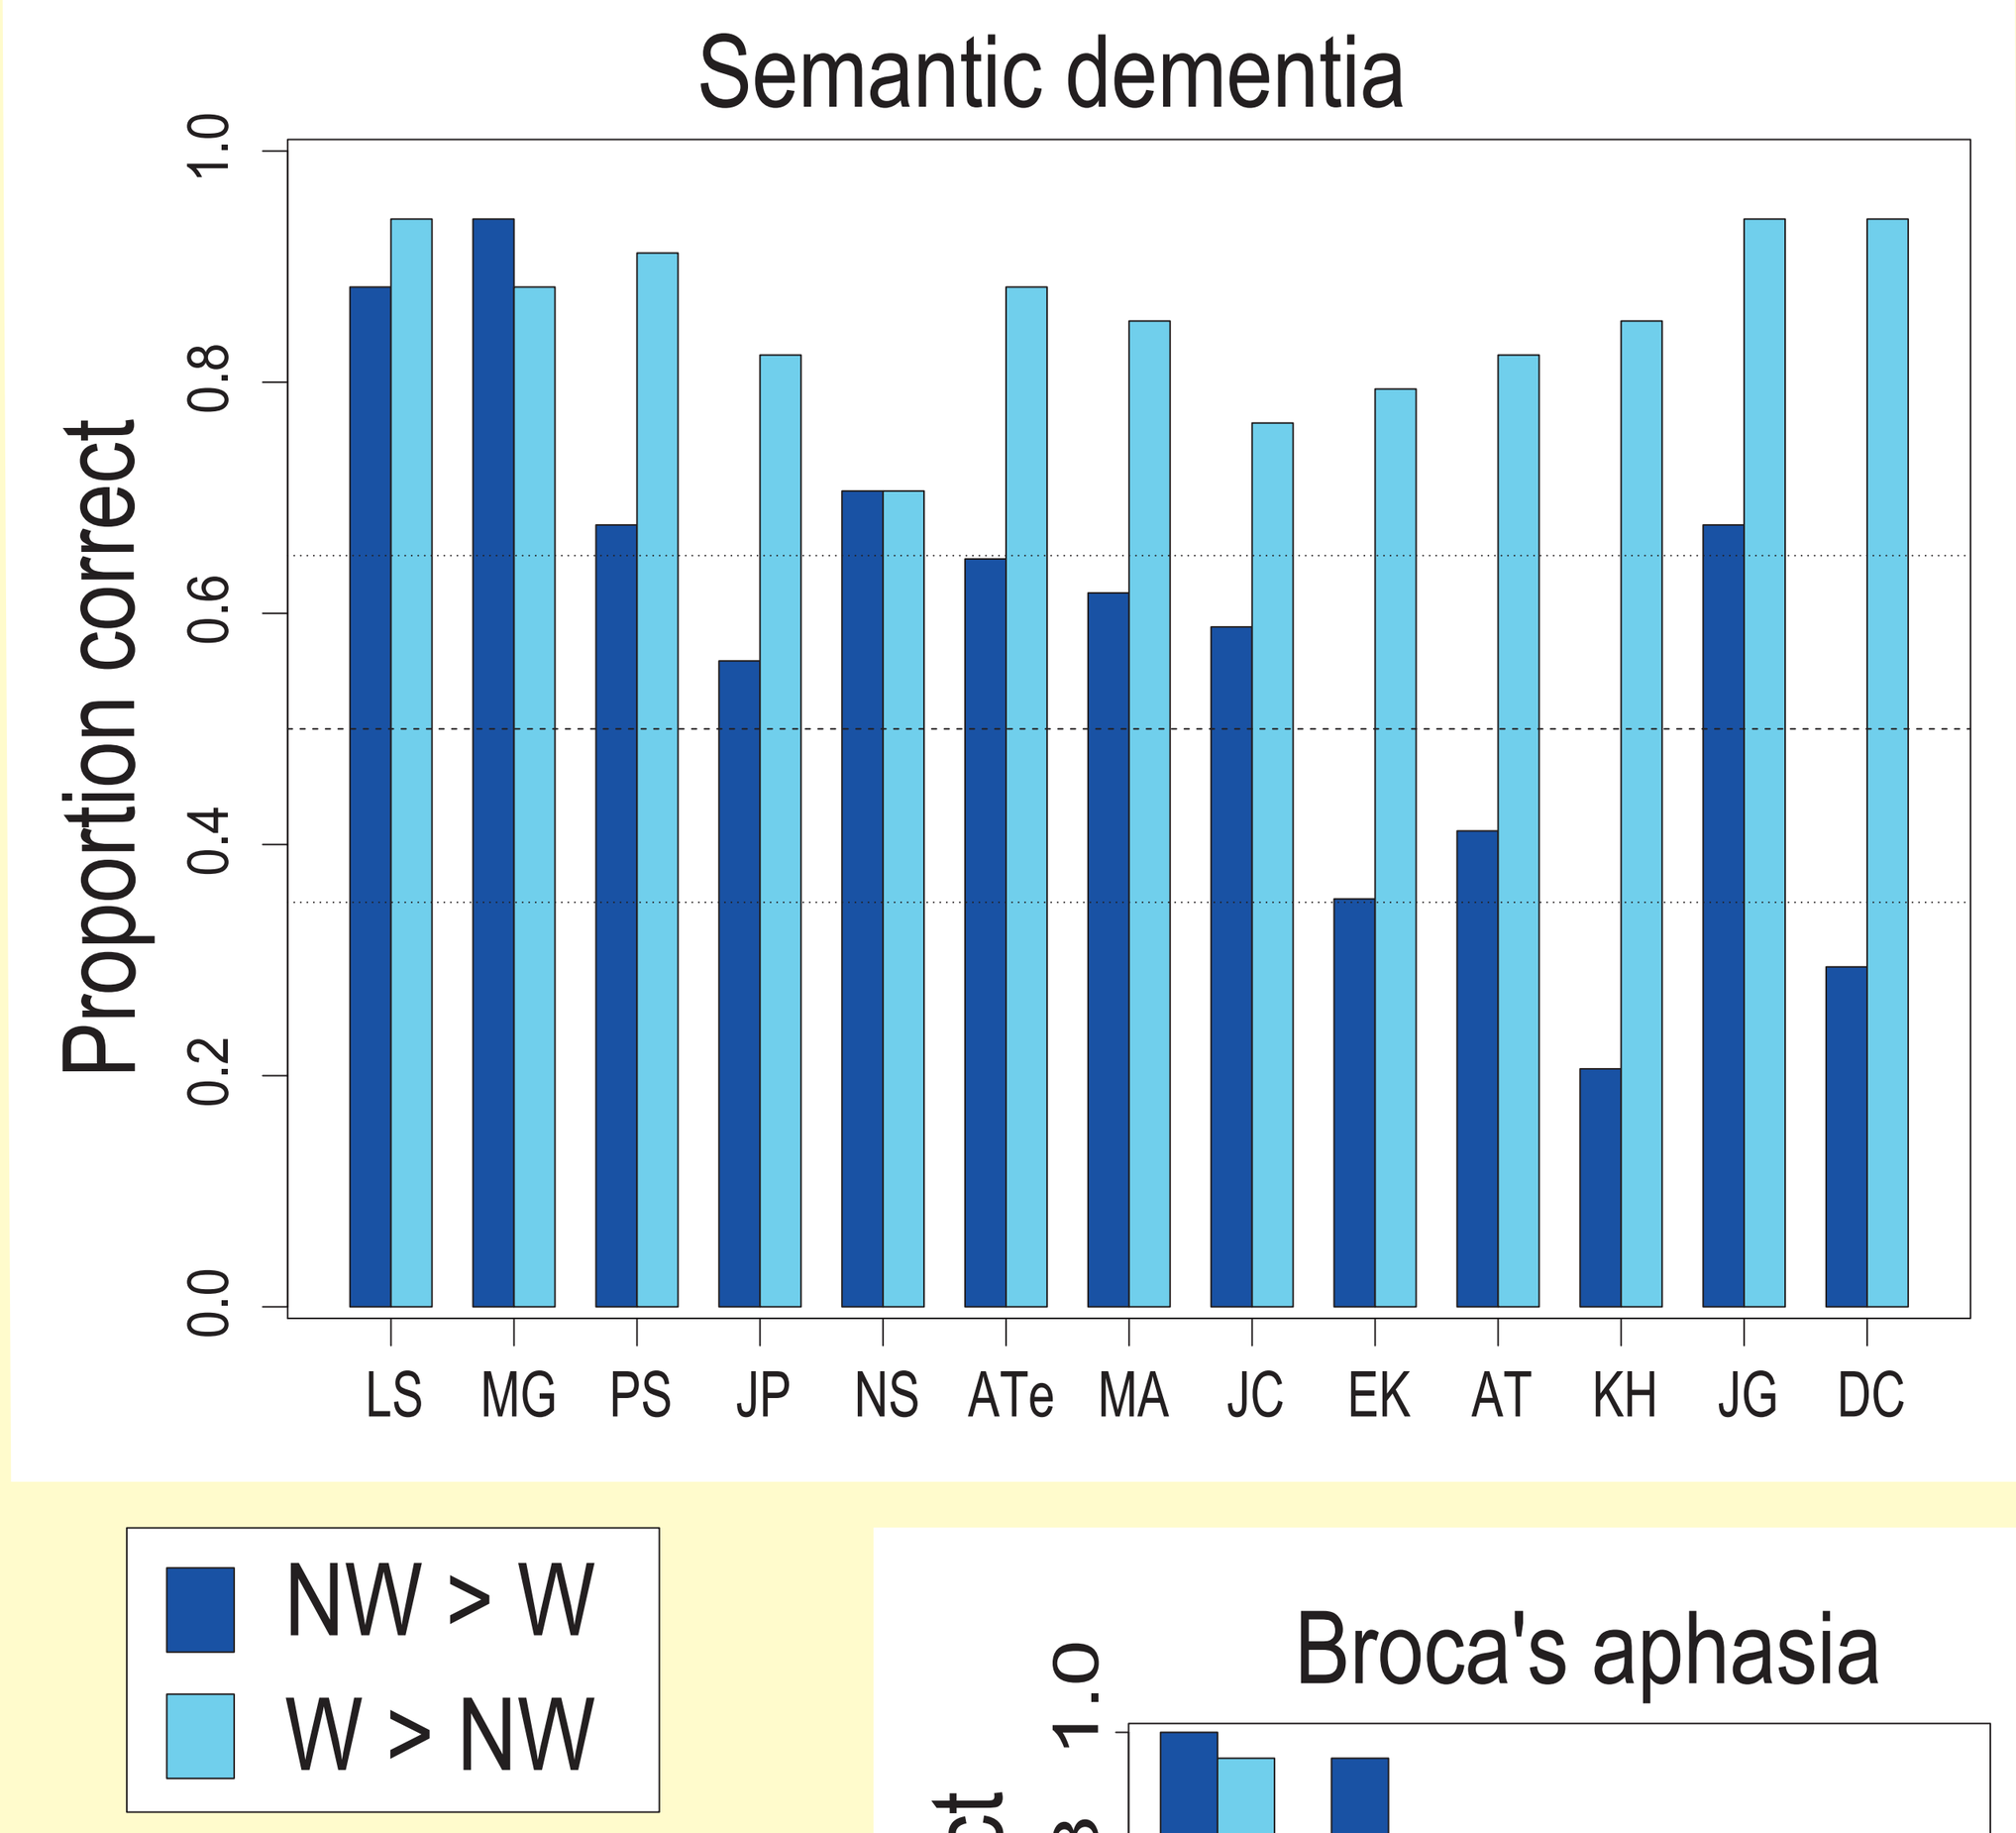
Semantic dementia
Proportion correct
LS MG PS JP NS ATe MA JC EK AT KH JG DC
NW > W
W > NW

■ Patients with semantic dementia show increasing impairment in NW > W condition and mild impairment in W > NW condition.

■ Broca's aphasics show comparable degrees of impairment in both conditions

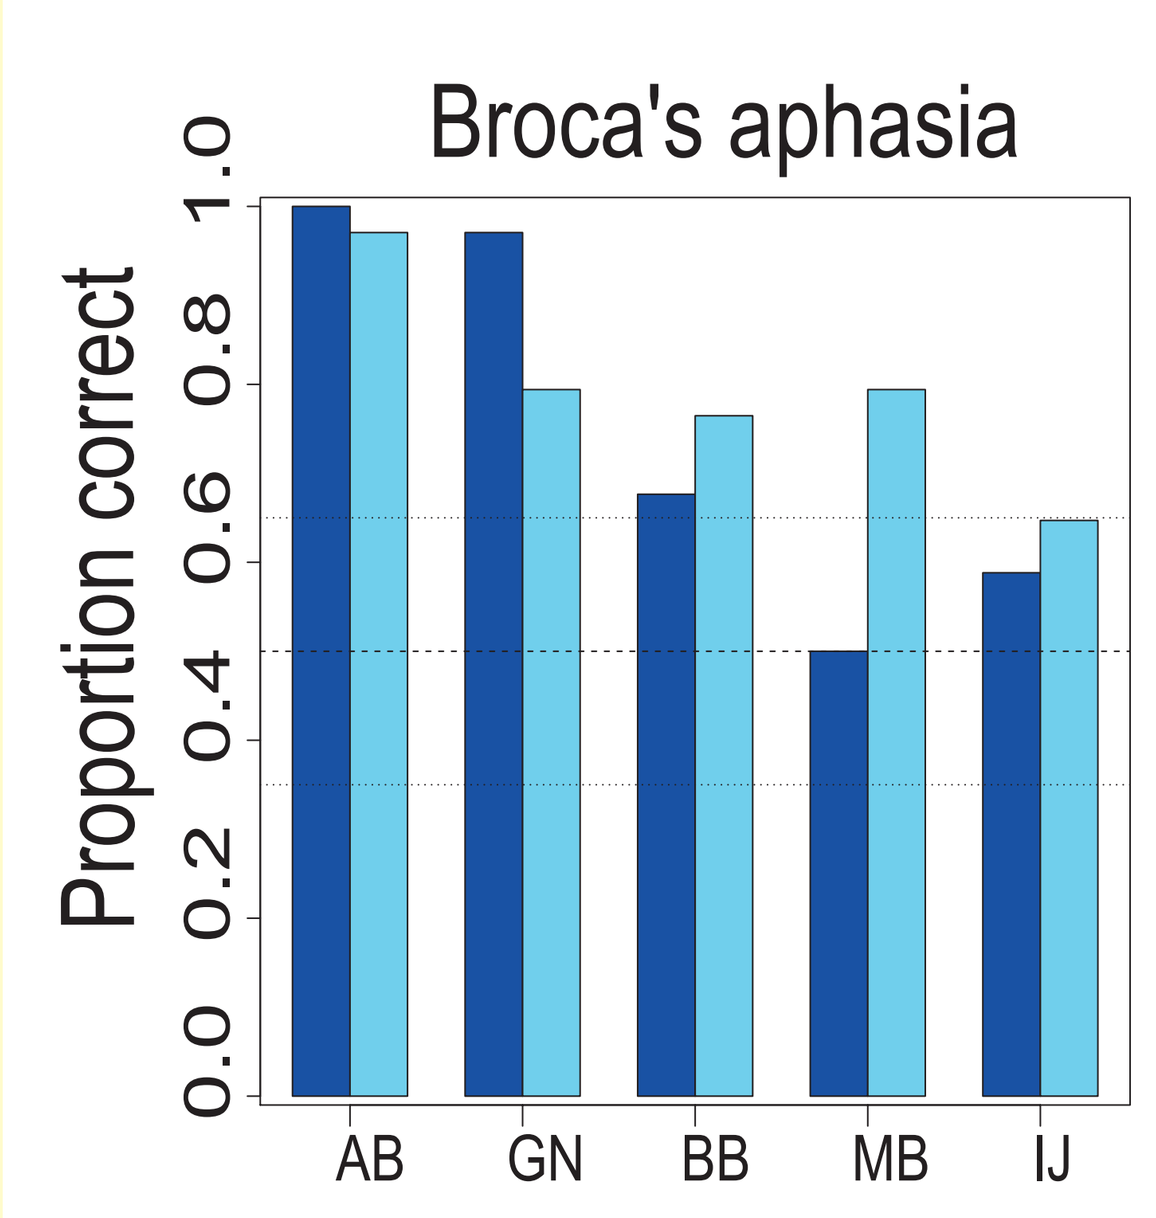
Broca's aphasia
Proportion correct
AB GN BB MB IJ

Patient means by stimulus type and word frequency

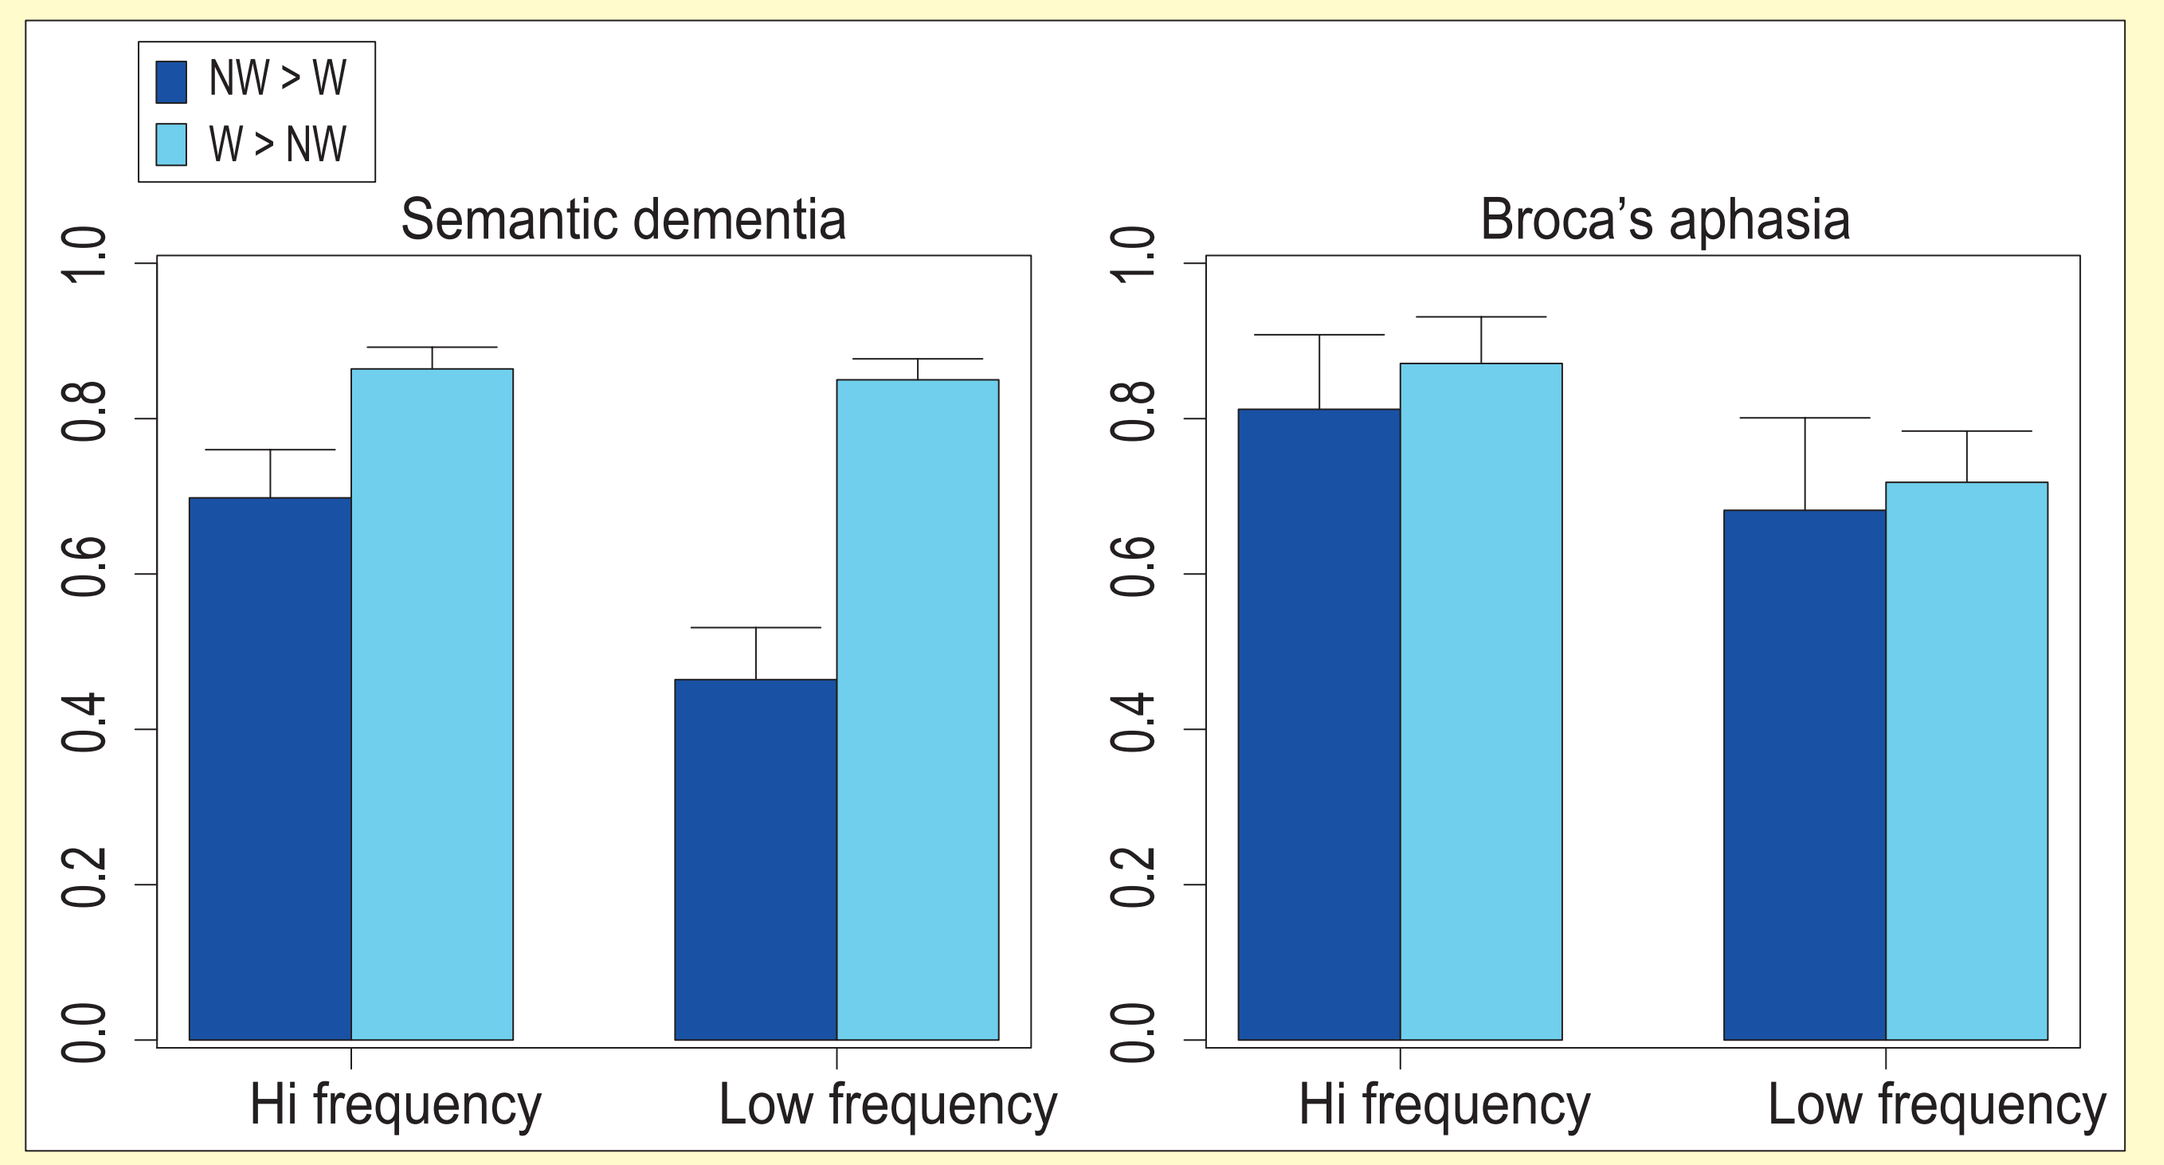
NW > W
W > NW
Semantic dementia
Broca's aphasia
Hi frequency
Low frequency

Repeated-measures GLM: Accuracy = WordFrq + StimType + (WF * ST)

Semantic dementia	Broca's aphasia
WordFrq: $F(1,12) = 25$, $p < 0.001$	WordFrq: $F(1,4) = 15$, $p < 0.02$
StimType: $F(1,12) = 21$, $p < 0.001$	StimType: $F(1,4) = 0.3$, p = n.s.
WF * ST: $F(1,12) = 9$, $p < 0.01$	WF * ST: $F(1,4) = 0.03$, p = n.s.

Implications for theories of object- and word- recognition

1. Patients with semantic dementia increasingly tend to accept "regular" stimuli and reject "irregular" stimuli in both object-decision and lexical-decision tasks, regardless of whether the "regular" item is a real object or a real word.
2. Severely impaired patients can perform well on either task when targets respect the regularities of the domain, and distractors do not. The apparent sparing of word or object recognition under semantic impairment may result from the structure of the testing materials used in the task, and not from the selective preservation of pre-semantic "structural descriptions" or lexical representations.
3. Targets and distractors in the standard object-decision task (the BORB) are not matched for typicality. Performance on this task falls between performance in the two conditions of the OAT.
4. Comparison of lexical decision in semantic dementia and Broca's aphasia suggests that the poor performance of SD patients in the NW>W condition was not simply due to this condition being harder.
5. The results are consistent with the predictions of distributed and recurrent theories of written-word and visual-semantic processing.

References

Humphreys, G. W., Riddoch, M. J. and Quinlan, P. T. (1988). Cascade processes in picture identification. Cognitive Neuropsychology, 5 (1), 67-103.

Coltheart, M., Rastle, K., Perry, C., Langdon, R., Ziegler, J. (2001). DRC: A dual-route cascaded model for visual word recognition and reading aloud. Psychological Review, 108 (1), 204-256.

Plaut, D. C., McClelland, J. L., Seidenberg, M., and Patterson, K. (1996). Understanding normal and impaired word reading: Computational principles in quasi-regular domains. Psychological Review, 103 (1), 56-115.

Rogers, T. T., Lambon Ralph, M. A., Patterson, K. McClelland, J. L., and Hodges, J. R. (1999). A recurrent connectionist model of semantic dementia. Poster presented at the 1999 meeting of the Cognitive Neuroscience Society.

* NOTE: M. A. Lambon Ralph contributed to this work while at the University of Bristol, and is now at the University of Manchester, Manchester UK.